64 AUGUST 2023 AUANEWS

AUA2023 BEST POSTERS

Off-the-Shelf Implant to Bridge a Urethra: Multicenter 8-Year Journey From Bench to Bed

Ganesh P. Vythlingam MBBS, MRCS, MSurg, FEAPU, PhD
University Malaya Medical Centre, Kuala Lumpur, Malaysia

Hans M. Larsson, PhD
Regenosca SA, Lausanne, Switzerland

Yeoh Wei Sien BMed Sci, MD, MSurg, FRCS (Urol) Glasgow
University Malaya Medical Centre, Kuala Lumpur, Malaysia

Saiful Azli bin Mohd Zainuddin MD, MSurg, FRCS (Urol) Glasgow
Ministry of Health, Kuala Lumpur, Malaysia

Eva-Maria Engelhardt, PhD
Regenosca S.A., Lausanne, Switzerland

Anand Sanmugam MD, MRCS, MSurg
University Malaya Medical Centre, Kuala Lumpur, Malaysia

Ong Teng Aik, MBBS, MSurg, FRCS (Urol) Glasgow
University Malaya Medical Centre, Kuala Lumpur, Malaysia

A surgically efficient and cost-effective tissue-engineered implant to reconstruct the urethra has not yet replaced current surgical practice of using autologous tissue grafts. The harvesting of these grafts is associated with complications at the donor site. Cell-based approaches have shown progress. However, these have been associated with high costs and logistical challenges, and have not replaced the current gold standard. An acellular, off-the-shelf implant with good regenerative potential is more likely to be used in clinical practice than a cellular implant.[1]

> "These new preclinical data further convinced the collaborating urologist to proceed with an application for a first-in-man clinical trial."

Though previously attempted, this has not shown efficacy to bridge large urethral defects and thus has failed to translate to clinical practice. We describe here our journey to achieve that objective in urethral reconstructive surgery. An important component for successful clinical translation was the establishment of early working collaboration between the bioengineers and urologist to design a clinically relevant implant. This multidisciplinary team was from different institutions based in Switzerland and Malaysia. Collagen was utilized as the foundation biomaterial of choice for the engineering of this envisioned urethral implant as it is a natural biomaterial and the most predominant protein in the human extracellular matrix. Multiple collagen implants were developed over a period of 8 years, all using animal-derived collagen as the base material. The various implant generations underwent in vitro testing, mechanical analyses, electron microscopy, and finally bench testing by urologist. In vivo testing was done in the rabbit urethral model to bridge a 2-cm defect with a tubular graft. In total, 69 male New Zealand rabbits were utilized to refine the final design, which was done both in Lausanne, Switzerland, and in Kuala Lumpur, Malaysia. Prior to euthanasia, all animals were subjected to a contrast voiding cystography to look for urethral patency, as done in clinical practice. Biopsies of repaired sites from euthanized rabbits were subjected to histology and immunohistochemistry. Tubular implants were then further tested in a clinically relevant defect of 4 cm in the dog model. First-generation implants did show promising regenerative potential, which we have published and presented.[2,3] Further improvement was done utilizing the data obtained to engineer an implant that had the best potential for clinical translation. The final prototype was implanted in 9 rabbits and 6 dogs. Rabbits were evaluated at 1, 3, and 6 months per our established protocol, and the dogs were examined by clinical patency assessment and histology up to 16 months. These new preclinical data further convinced the collaborating urologist to proceed with an application for a first-in-man clinical trial. The incorporation of Regenosca SA in Switzerland in 2019 initiated the transition from a research product developed in an academic setting to a clinical-grade product developed and produced according to regulatory standards (Figure 1).

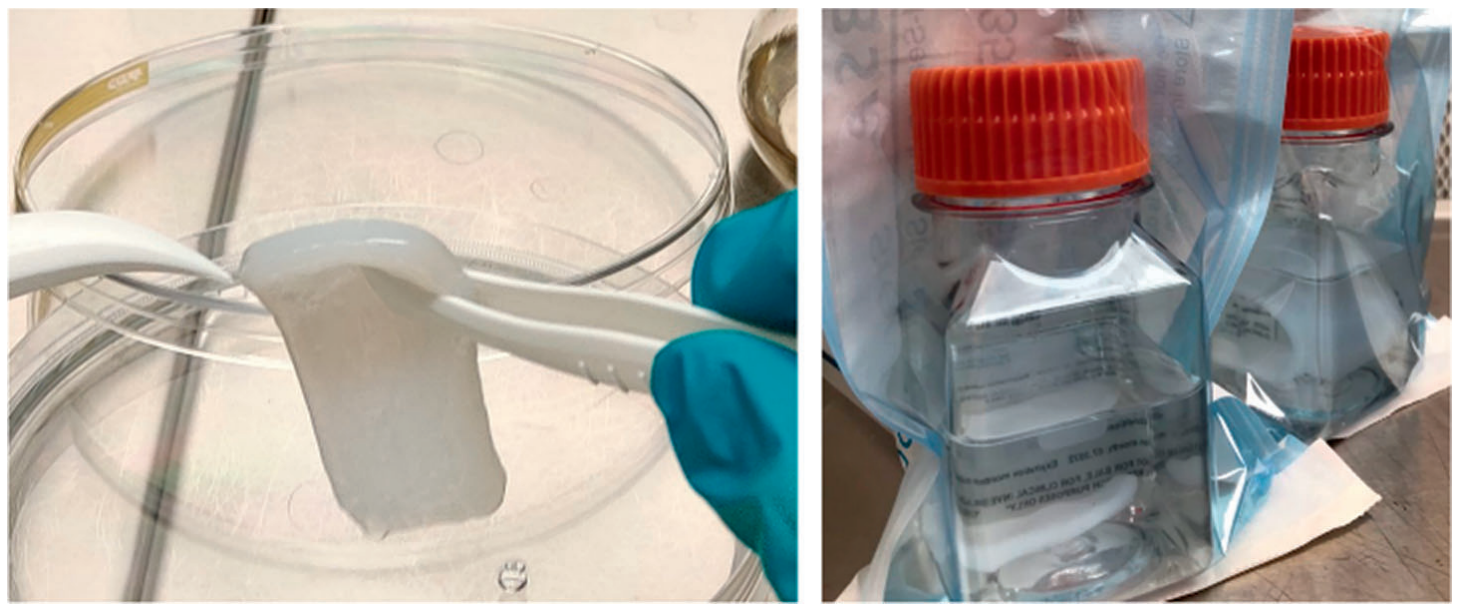

Figure 1. Off-the-shelf available implant manufactured in a Good Manufacturing Practice facility for urethral reconstruction.

Approval was subsequently obtained from the Medical Research Ethics Committee at University of Malaya Medical Centre, Malaysia in 2020, for a 5-patient first-in-man study to evaluate the implants for substitution urethroplasty of distal urethral strictures of 3 cm. Following the pandemic, the first patients were operated on in August 2022 using the acellular collagen implant manufactured with Regenosca's TissueSpan technology, employing standard surgical technique (Figure 2).

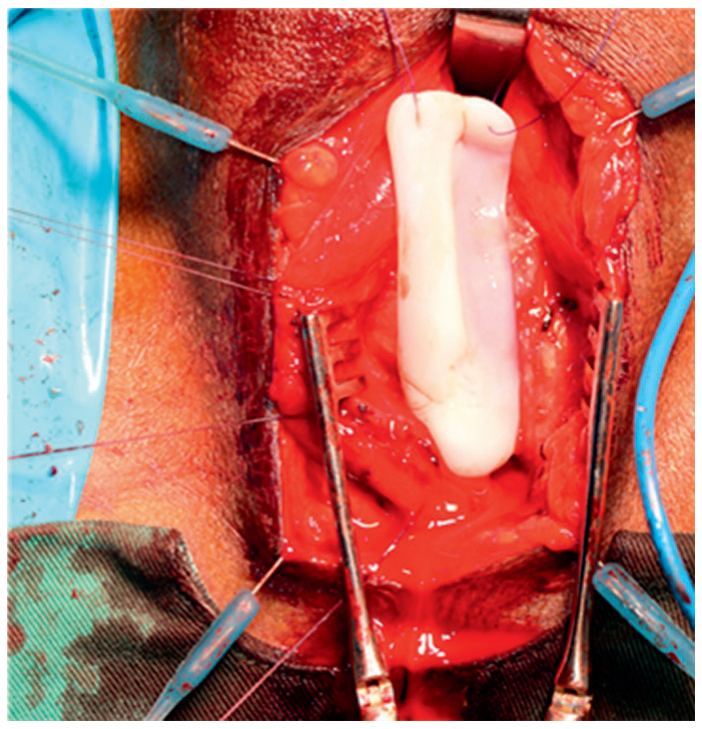

Figure 2. TissueSpan implant utilized in the first-in-man urethral reconstruction study in August 2022.

Currently, 2 patients have been implanted with the Regenosca product bridging urethral defects of less than 3 cm. The first 2 patients showed good clinical outcome and we have initiated patient recruitment for the remaining 3 surgeries. The acellular implant may have the potential to be an off-the-shelf product for substitution urethroplasty. Its mechanical properties allow surgeons to easily recreate a physical conduit while its material properties favor tissue regeneration in the long term. A manuscript is currently in preparation to show the preclinical and clinical data. Large-scale clinical trials, however, are required to further confirm safety, performance, and clinical benefit to patients. ■

1. Pastorek D, Culenova M, Csobonyeiova M, Skuciova V, Danisovic L, Ziaran S. Tissue engineering of the urethra: from bench to bedside. *Biomedicines.* 2021;9(12):1917.
2. Pinnagoda K, Larsson HM, Vythilingam G, et al. Engineered acellular collagen scaffold for endogenous cell guidance, a novel approach in urethral regeneration. *Acta Biomaterialia.* 2016;43:208-217.
3. Larsson HM, Vythilingam G, Pinnagoda K, et al. Fiber density of collagen grafts impacts rabbit urethral regeneration. *Sci Rep.* 2018;8(1):10057.